P2.08-07

EDGE-Lung: A Phase 2 Open-Label Platform Study to Evaluate Immunotherapy-Based Combinations in Patients with Metastatic NSCLC

Melissa Johnson,[1] Arvind Chaudhry,[2] William Fox,[3] Sasha Han,[4] Christoph M. Ahlers,[4] Trever Todd,[4] Joseph K. Park,[5] Aparna Hegde[6]

[1]Sarah Cannon Research Institute, Nashville, TN, USA; [2]Medical Oncology Associates, Spokane, WA, USA; [3]Mid North Coast Cancer Institute, Coffs Harbour, NSW, Australia; [4]Arcus Biosciences, Hayward, CA, USA; [5]Gilead Sciences, Foster City, CA, USA; [6]O'Neal Comprehensive Cancer Center, University of Alabama, Birmingham, AL, USA

BACKGROUND

- Immunotherapy agents targeting the programmed cell death protein 1 (PD-1)/programmed death ligand 1 (PD-L1) axis have improved outcomes for patients with locally advanced or metastatic non–small cell lung cancer (NSCLC)
- Despite these advances in NSCLC management, many patients eventually progress, and long-term survival rates remain low;[1-3] thus, there is an urgent need for improved treatment options
- Novel treatment regimens combining anti–PD-[L]1 therapy with additional immune checkpoint inhibitors may provide increased clinical benefit over anti–PD-[L]1 monotherapy in patients with NSCLC[1,4,5]
 - Interim results from the ARC-7 study suggested that the combination of domvanalimab and zimberelimab may provide a clinically meaningful benefit with a manageable safety profile in patients with NSCLC, compared to zimberelimab (anti–PD-1) monotherapy[6]
 - Preliminary results from ARC-8 demonstrated encouraging activity with quemliclustat in combination with standard of care chemotherapy and zimberelimab in patients with metastatic pancreatic adenocarcinoma[7]

Investigational Therapies

- Quemliclustat (AB680) is a potent, selective, small molecule inhibitor of CD73; by blocking production of adenosine through CD73 inhibition, quemliclustat may help reverse tumor-directed immunosuppression, activating host antitumor immunity (**Figure 1**)[8]
- Domvanalimab (AB154) is an Fc-silent, humanized, immunoglobulin G1 monoclonal antibody that blocks the binding of the checkpoint receptor T cell immunoglobulin and ITIM domain (TIGIT) to its ligand CD155, reducing inhibition of T cells and natural killer (NK) cells and promoting antitumor activity (**Figure 2**)[9-12]
 - Blocking the CD155-TIGIT interaction allows the immunostimulatory binding of CD155 to CD226
 - As domvanalimab is Fc-silent, it does not stimulate antibody-dependent cellular cytotoxicity-mediated destruction of TIGIT-bearing immune cells
- Zimberelimab (AB122) is a fully human IgG4 monoclonal antibody that binds PD-1 on T cells and NK cells, preventing PD-L1–mediated immunosuppressive effects and resulting in enhanced immune-mediated tumor cell death (**Figure 2**)[13,14]

Figure 1. Targeting the Adenosine Pathway

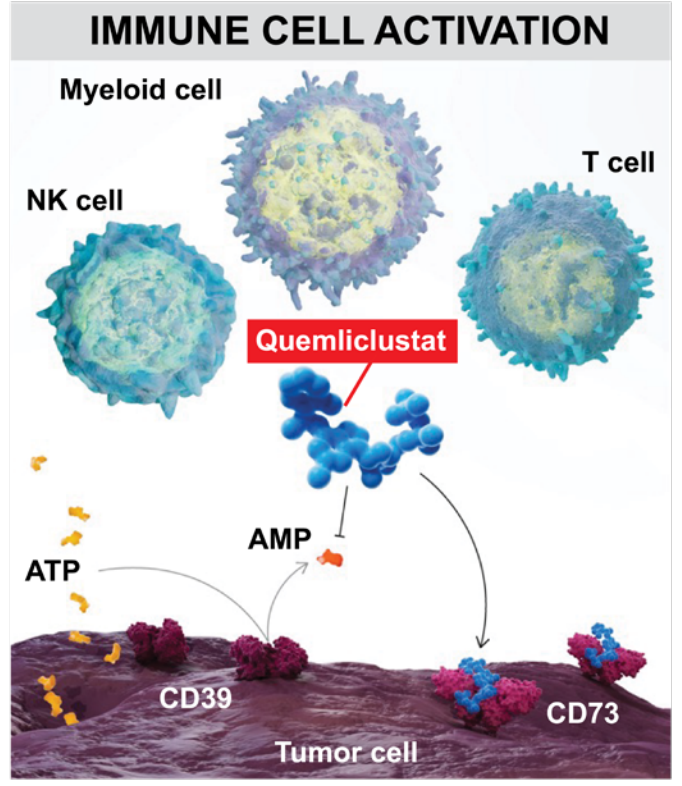
IMMUNE CELL ACTIVATION
Myeloid cell
NK cell
T cell
Quemliclustat
ATP
AMP
CD39
CD73
Tumor cell

AMP, adenosine monophosphate; ATP, adenosine triphosphate; NK, natural killer.

Figure 2. Checkpoint Inhibition and the TIGIT Pathway

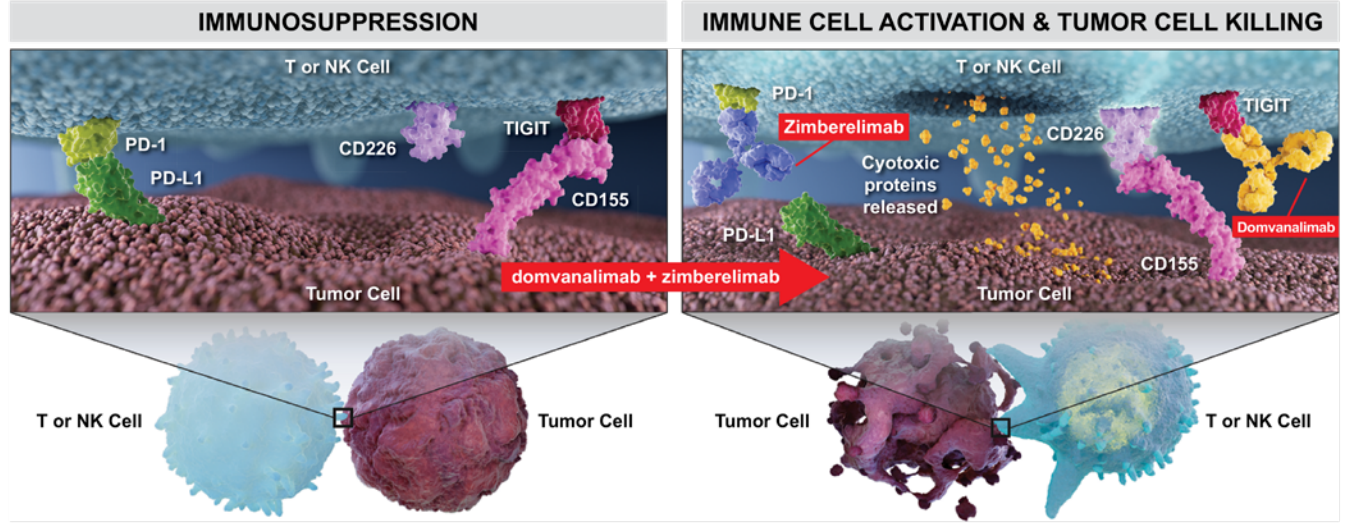
IMMUNOSUPPRESSION
IMMUNE CELL ACTIVATION & TUMOR CELL KILLING
T or NK Cell
PD-1
PD-L1
CD226
TIGIT
CD155
Tumor Cell
domvanalimab + zimberelimab
Zimberelimab
Cytoxic proteins released
Domvanalimab
T or NK Cell
Tumor Cell
Tumor Cell
T or NK Cell

PD-1, programmed cell death protein 1; PD-L1, programmed cell death ligand 1; NK, natural killer; TIGIT, T cell immunoglobulin and ITIM domain.

Study Objective

- The EDGE-Lung study will investigate the safety and clinical activity of novel quemliclustat- and domvanalimab-based treatment combinations in patients with metastatic NSCLC

Study Design

- EDGE-Lung (NCT05676931) is a global, multicenter, randomized, open-label, 2-stage platform, phase 2 trial (**Figure 3**)
- Different NSCLC patient populations will be treated with novel regimens via separate sub-studies. A minimum of approximately 190 patients, and up to approximately 320 patients, will be enrolled
 - Each sub-study will enroll 20 to 30 patients per treatment arm during the preliminary stage; cohorts that meet prespecified criteria may enroll further in the expansion phase, up to a total of 40 patients per treatment arm
 - Within each sub-study, patients will be randomly assigned equally between each arm open at the time of enrollment
 - If a patient is eligible for more than 1 enrolling sub-study, the investigator will determine which sub-study the patient will be enrolled into
 - Treatment arms with quemliclustat will include a dose-escalation safety run-in following a standard 3 + 3 design
- This platform study is designed with flexibility to explore additional study treatment regimens within a sub-study, stop accrual into existing treatment regimens that demonstrate minimal clinical activity or unacceptable toxicity, or explore additional NSCLC population(s) with 1 or more new sub-studies via protocol amendment

Figure 3. EDGE-Lung Study Design

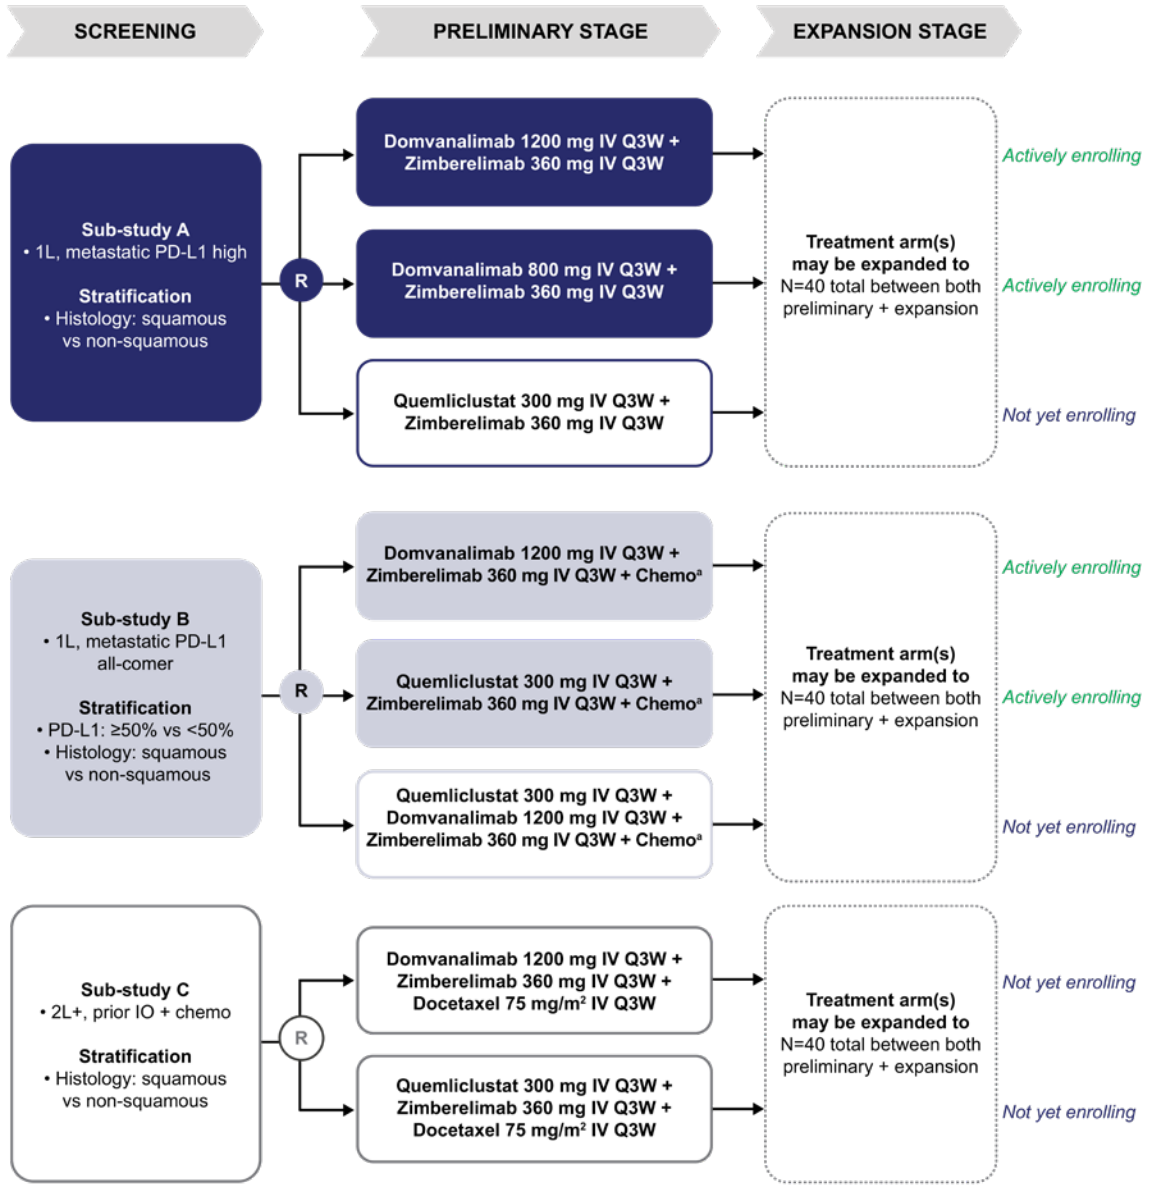
SCREENING
PRELIMINARY STAGE
EXPANSION STAGE
Sub-study A
• 1L, metastatic PD-L1 high
Stratification
• Histology: squamous vs non-squamous
R
Domvanalimab 1200 mg IV Q3W + Zimberelimab 360 mg IV Q3W
Domvanalimab 800 mg IV Q3W + Zimberelimab 360 mg IV Q3W
Quemliclustat 300 mg IV Q3W + Zimberelimab 360 mg IV Q3W
Treatment arm(s) may be expanded to N=40 total between both preliminary + expansion
Actively enrolling
Actively enrolling
Not yet enrolling
Sub-study B
• 1L, metastatic PD-L1 all-comer
Stratification
• PD-L1: ≥50% vs <50%
• Histology: squamous vs non-squamous
R
Domvanalimab 1200 mg IV Q3W + Zimberelimab 360 mg IV Q3W + Chemo[a]
Quemliclustat 300 mg IV Q3W + Zimberelimab 360 mg IV Q3W + Chemo[a]
Quemliclustat 300 mg IV Q3W + Domvanalimab 1200 mg IV Q3W + Zimberelimab 360 mg IV Q3W + Chemo[a]
Treatment arm(s) may be expanded to N=40 total between both preliminary + expansion
Actively enrolling
Actively enrolling
Not yet enrolling
Sub-study C
• 2L+, prior IO + chemo
Stratification
• Histology: squamous vs non-squamous
R
Domvanalimab 1200 mg IV Q3W + Zimberelimab 360 mg IV Q3W + Docetaxel 75 mg/m² IV Q3W
Quemliclustat 300 mg IV Q3W + Zimberelimab 360 mg IV Q3W + Docetaxel 75 mg/m² IV Q3W
Treatment arm(s) may be expanded to N=40 total between both preliminary + expansion
Not yet enrolling
Not yet enrolling

1L, first line; 2L+, second or greater line; Chemo, chemotherapy; IO, immuno-oncology therapy; IV, intravenous; Q3W, every 3 weeks.
[a]Platinum doublet chemotherapy.

METHODS

Patient Population

- Eligible patients are adults with histologically confirmed metastatic squamous or non-squamous NSCLC
 - Eligible treatment-naive patients with high PD-L1 expression may be enrolled in either sub-study A or sub-study B at the discretion of the investigator
- Key inclusion and exclusion criteria are shown in **Table 1**

Table 1. Key Inclusion and Exclusion Criteria

Key Inclusion Criteria	Key Exclusion Criteria
General inclusion criteria • Age ≥18 years • Histologically-confirmed stage IV squamous or non-squamous NSCLC per AJCC version 8 • ECOG performance status of 0 or 1 • ≥1 measurable target lesion per RECIST v1.1	**General exclusion criteria** • Presence of *ALK* fusion oncogene or actionable *EGFR* mutation[a] • Known presence of any other genomic aberration or driver mutation (e.g. *ROS, BRAF, NTRK*) for which targeted therapy is locally available
Sub-study A–specific inclusion criteria • Treatment-naive for metastatic disease • Previously documented high PD-L1 expression (TPS ≥ 50% by PharmDx 22C3 or 28-8 PharmDx [Dako], or TC ≥50% by SP263 [Ventana])	**Sub-study A–specific exclusion criteria** • Prior treatment in the metastatic setting with any anti–PD-[L]1 antibody or any other therapeutic antibody targeting an immune checkpoint
Sub-study B–specific inclusion criteria • Treatment-naive for metastatic disease • Previously documented PD-L1 expression (by PharmDx 22C3 or 28-8 PharmDx [Dako], or by SP263 [Ventana]) • CrCl ≥45 mL/min (60 mL/min for patients receiving cisplatin) by Cockroft-Gault equation	**Sub-study B–specific exclusion criteria** • Prior treatment in the metastatic setting with any anti–PD-[L]1 antibody or other therapeutic antibody targeting an immune checkpoint
Sub-study C-specific inclusion criteria • PD or recurrence after platinum-based chemotherapy and anti–PD-[L]1 therapy, given concurrently or sequentially • No PD for at least 12 weeks after initiation of prior anti–PD-[L]1 therapy • Documented radiographic PD on or after the most recent regimen for metastatic NSCLC • No more than 2 prior lines of systemic therapy for metastatic disease	**Sub-study C-specific exclusion criteria** • Prior lung cancer treatment with docetaxel, anti-TIGIT, or anti-adenosine therapies

AJCC, American Joint Committee on Cancer staging; CrCl, creatinine clearance; ECOG, Eastern Cooperative Oncology Group; NSCLC, non–small cell lung cancer; PD, progressive disease; PD-L1, programmed cell death ligand 1; RECIST, Response Evaluation Criteria in Solid Tumors; TC, tumor cell; TIGIT, T cell immunoglobulin and ITIM domain; TPS, tumor proportion score.
[a]Testing is mandatory for patients with non-squamous histology and optional for those with squamous histology.

Outcomes and Endpoints

- Treatment will be continued until unacceptable toxicity, loss to follow-up, disease progression, study withdrawal, death, or study termination
- Key study endpoints are listed in **Table 2**

Table 2. Key Study Endpoints

Primary Endpoints	Secondary Endpoints	Exploratory Endpoints
• Objective response rate[a] • Safety	• Disease control rate[a] • Progression-free survival[a] • Duration of response[a] • Overall survival • Pharmacokinetics • Immunogenicity	• Biomarkers • Pharmacodynamics

RECIST, Response Evaluation Criteria in Solid Tumors.
[a]Assessed by the investigator according to RECIST v1.1.

Statistical Considerations

- Efficacy and safety will be analyzed for all enrolled patients who received any amount of study treatment
- For quemliclustat-containing arms that include a safety run-in, patients will be evaluable for the safety run-in phase if they have received at least 1 dose of treatment and completed safety follow-up assessments for at least 1 full 21-day treatment cycle

Status

- The study is currently planned to enroll patients in Asia-Pacific, Europe, and North America

REFERENCES

1. Xia L et al. *Oncologist*. 2019;24:S31-S41.
2. de Castro G et al. *J Clin Oncol*. 2023:41:1986-1991.
3. Reck M et al. *J Clin Oncol*. 2021;39:2339-2349.
4. Marei HE et al. *Cancer Cell Int*. 2023;23:64.
5. Rocco D et al. *Expert Opin Biol Ther*. 2023;23:261-268.
6. Johnson M et al. Presented at: ASCO Plenary Series; December 20, 2022; Virtual.
7. Manji GA et al. ASCO 2021. Abstract #404.
8. Piovesan D et al. *Mol Cancer Ther*. 2022;21:948-959.
9. Le Mercier I et al. *Front Immunol*. 2015;6:1-15.
10. Martinet L, Smyth MJ. *Nat Rev Immunol*. 2015;15:243-254.
11. Johnston RJ et al. *Cancer Cell*. 2014;26:923-937.
12. Zhang Q et al. *Nat Immunol*. 2018;19:723-732.
13. Markam A. *Drugs*. 2021;81:2063-2068.
14. Yi M et al. *Mol Cancer*. 2022;21:28.

ACKNOWLEDGMENTS

The authors gratefully acknowledge the patients, their families, and their caregivers for their participation. The authors wish to thank Lauren Sharrer for her contributions to this study. This study was sponsored by Arcus Biosciences (Hayward, CA, USA) in collaboration with Gilead Sciences, Inc (Foster City, CA, USA). Medical writing assistance was provided by MEDiSTRAVA (San Diego, CA, USA) and funded by Arcus Biosciences and Gilead Sciences.

Author for correspondence: Melissa Johnson, MD; mjohnson@tnonc.com

Presented at the 2023 World Conference on Lung Cancer (WCLC); September 9-12, 2023; Singapore

Scan this QR Code to download the full poster

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from WCLC or the author of this poster.